AACR 2022
2066

In vitro and in vivo properties of MDK-703: An Fc-peptide fusion IL-7Rαγc agonist unrelated in structure to IL-7

Angie I. Park, Steven E. Cwirla, Alice V. Bakker, Blake M. Williams, Prarthana Joshi, Praechompoo Pongtornpipat, Sandra M. Wang, Bryan Baxter, Michael C. Needels, Ronald W. Barrett, William J. Dower. Medikine, Inc. Menlo Park, CA

Medikine

Introduction

IL-7 receptor (IL-7R) activation is essential for the proper development and homeostasis of T-cell subpopulations and maintenance of the TCR clonal repertoire. Emerging evidence indicates the potential clinical utility of IL-7R agonists for immunotherapy of lymphopenia, oncology, and other indications. However, IL-7-based proteins that have been studied in humans show a propensity to induce anti-drug antibodies (ADAs), including those that neutralize natural IL-7[1]. Here we report the discovery of MDK1472, a small novel peptidyl agonist of IL-7R with a MW <5000Da. This peptide is structurally unrelated to IL-7 and has a sequence that makes MDK1472 unlikely to generate IL-7 neutralizing antibodies. MDK1472 is fused to an IgG Fc-domain to improve pharmacokinetics and this fusion, MDK-703, exhibits biological properties similar to those of IL-7 *in vitro* and when administered to animals.

MDK1472 and MDK-703

MDK1472

- Synthetic peptide with sequence unrelated to IL-7 or other human proteins
- Binds to IL-7Rα and Rγc subunits and activates IL-7R with sub-nanomolar potency and full efficacy in pSTAT5 assay
- Rγc-binding portion of the molecule does not block the activity of IL-2, IL-4, IL-9, IL-15, or IL-21[2]
- In silico assessment of potential immunogenicity (EPIVAX) indicated a low probability of the presence of Class II (HLA-DR) restricted HLA ligands and putative T cell epitopes[3]

Fc fusion MDK-703 (Development lead candidate)

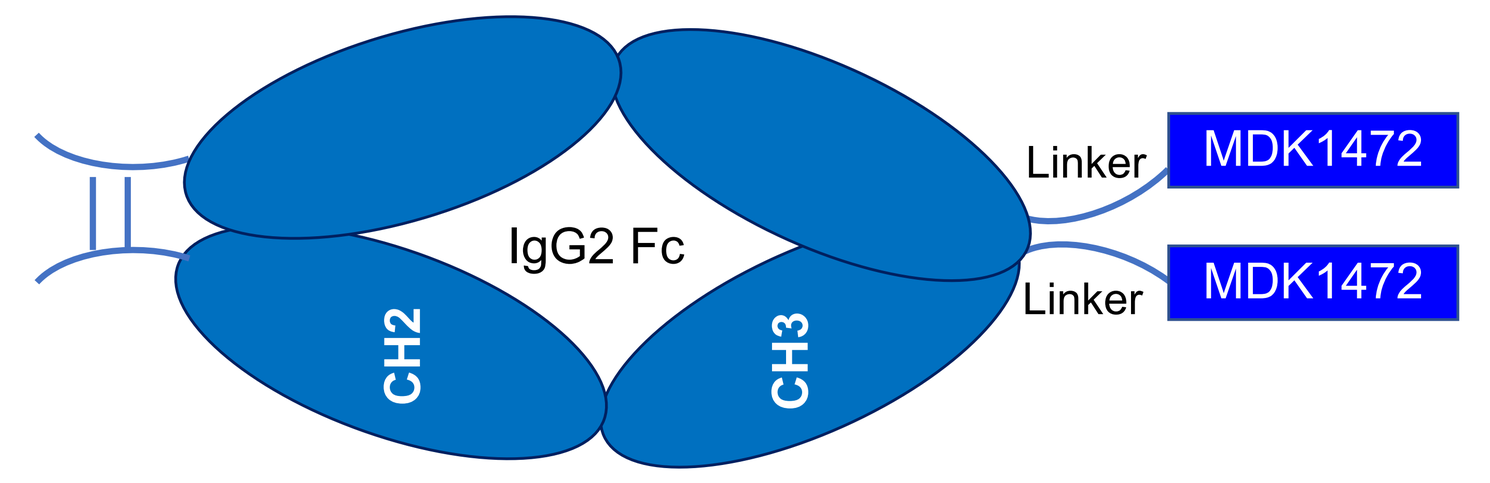
IgG2 Fc
CH2
CH3
Linker
MDK1472
Linker
MDK1472

MDK-703 Binding to IL-7Rα and Rγc

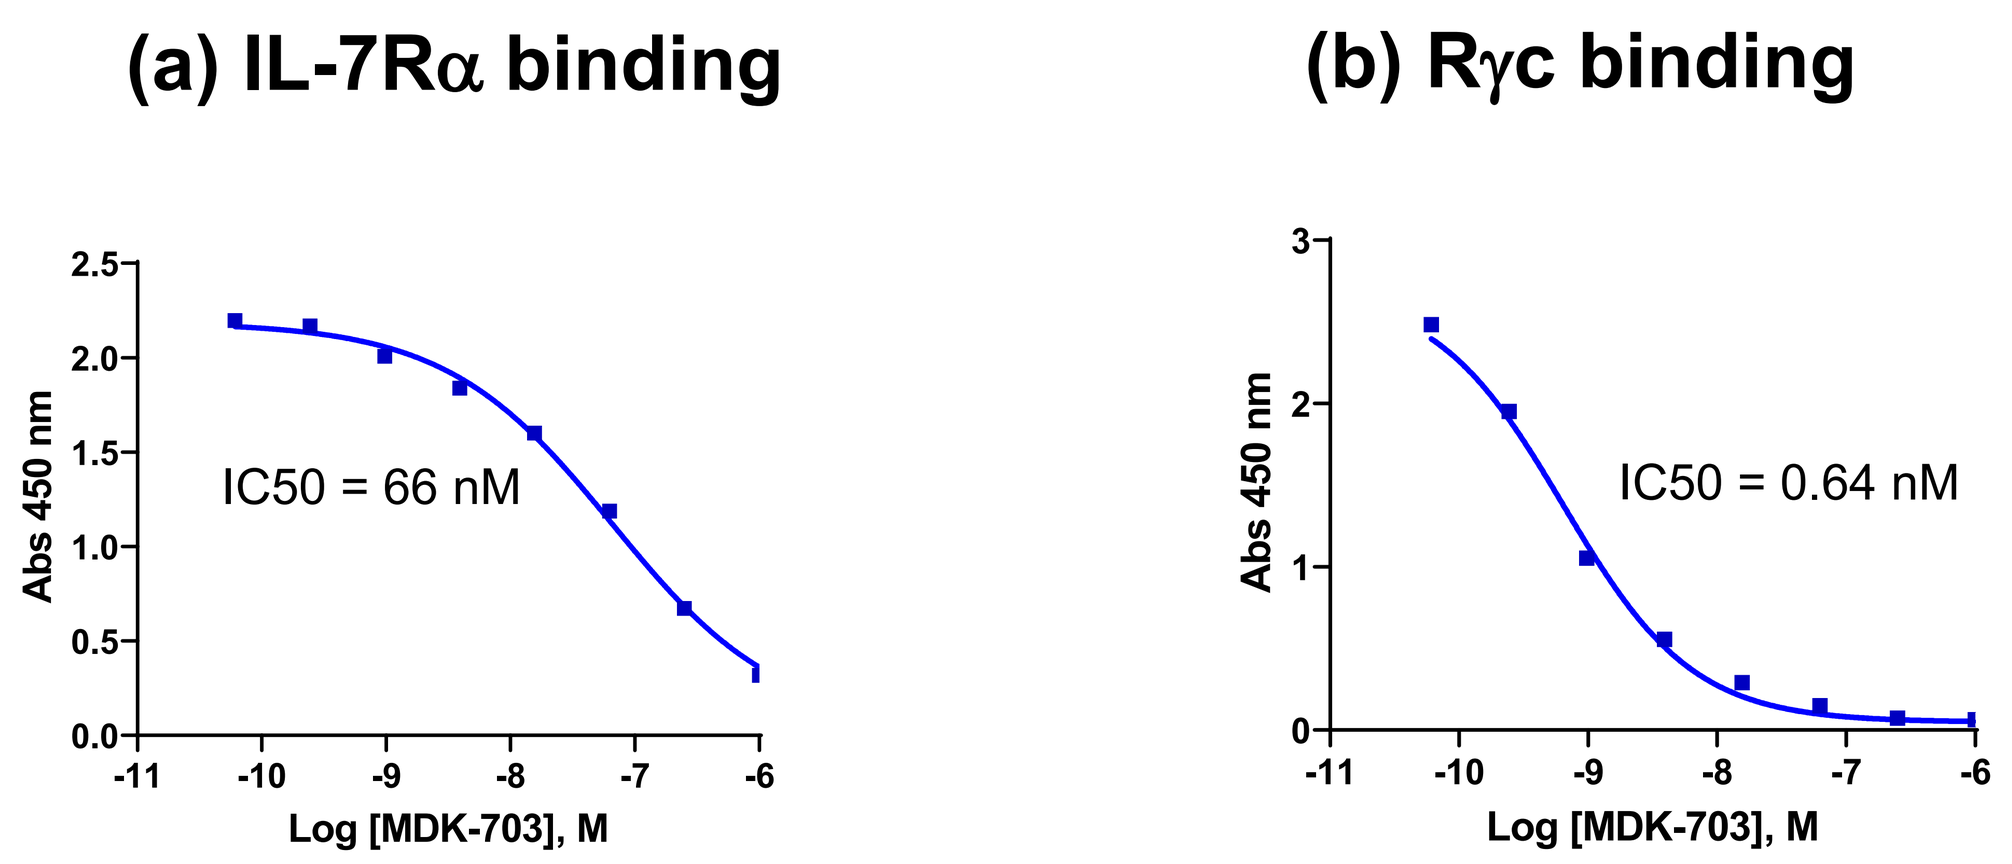
(a) IL-7Rα binding
IC50 = 66 nM
Abs 450 nm
Log [MDK-703], M
(b) Rγc binding
IC50 = 0.64 nM
Abs 450 nm
Log [MDK-703], M

Fig 1. MDK-703 binding to IL-7R subunits

The binding affinity of MDK-703 to IL-7Rα (**a**) and Rγc (**b**) was measured by competition ELISA. Serially diluted MDK-703 was added to plate wells coated with human IL-7Rα-Fc (**a**) or Rγc-Fc (**b**). After 1 hour, NeutrAvidin™-HRP:biotin-IL-7Rα binding peptide complex (**a**) or NeutrAvidin™-HRP:biotin-γc binding peptide complex (**b**) was added and incubated for 45 min. Bound complexes were quantified by measuring HRP activity using TMB substrate. IC50 values were generated using GraphPad Prism software.

MDK-703 Induces pSTAT5 in PBMCs

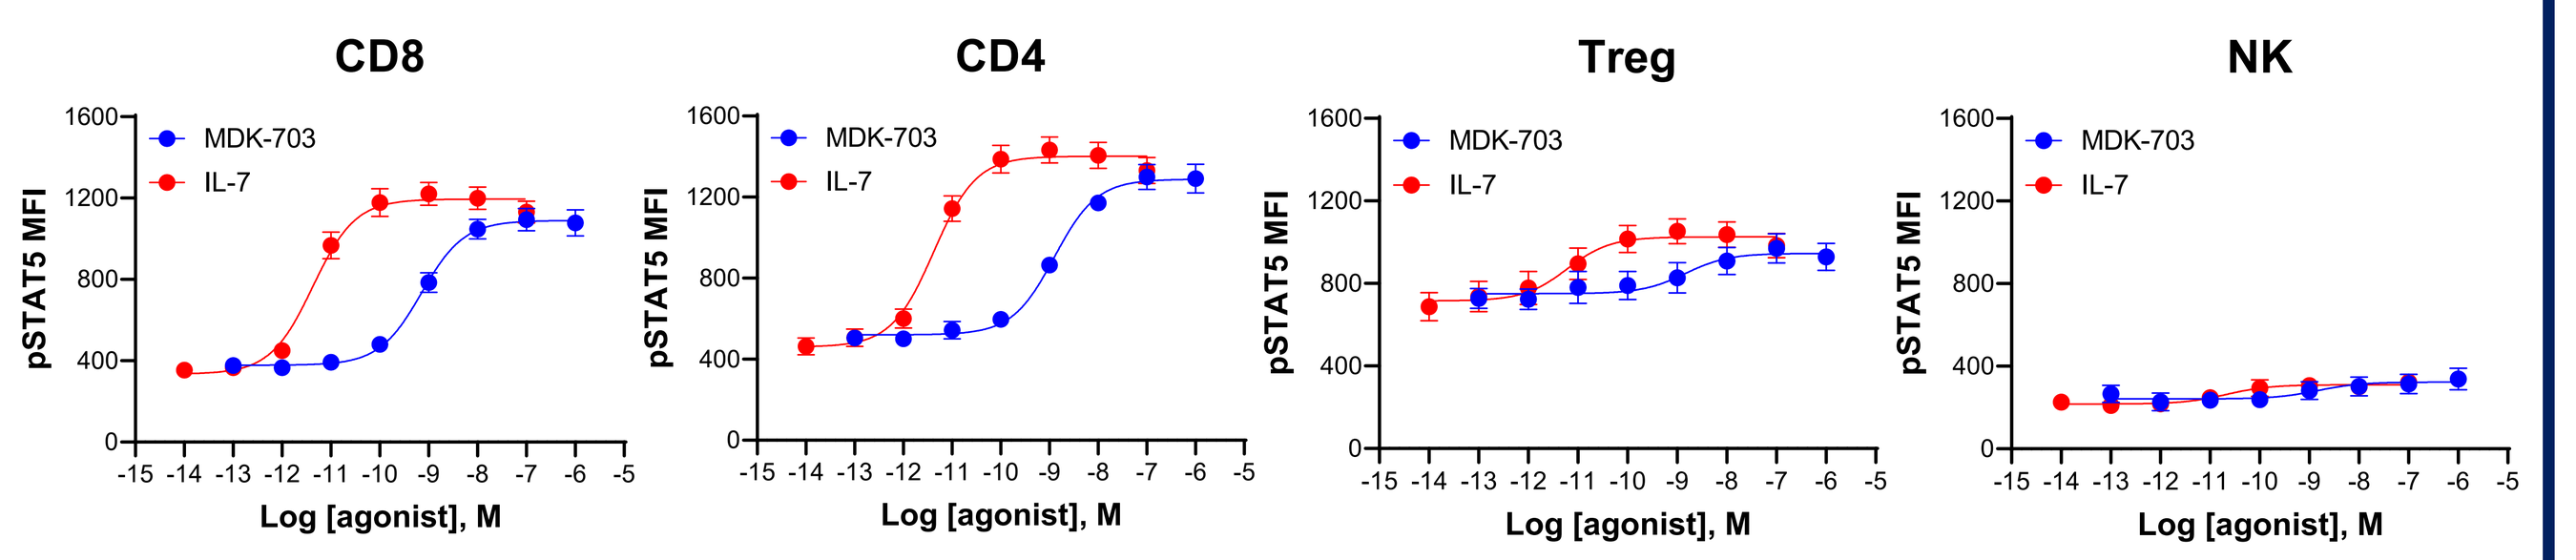
CD8
CD4
Treg
NK
MDK-703
IL-7
pSTAT5 MFI
Log [agonist], M

CD8

EC50	MDK-703	IL-7
Average	7.80E-10	4.40E-12
SEM	1.50E-11	3.70E-14

CD4

EC50	MDK-703	IL-7
Average	7.80E-10	4.40E-12
SEM	1.50E-11	3.70E-14

Treg

EC50	MDK-703	IL-7
Average	1.57E-09	6.27E-12
SEM	1.50E-11	3.70E-14

NK

EC50	MDK-703	IL-7
Average	1.54E-09	2.41E-11
SEM	3.05E-10	7.10E-12

Fig 2. Induction of pSTAT5 in PBMC treated with MDK-703 or IL-7. Frozen PBMCs from 5 healthy donors were rested overnight and stained for viability, followed by cell surface antibody staining on ice. Cells were washed and incubated with MDK-703 or IL-7 for 30 min at 37ºC. After washing, cells were fixed, permeabilized, stained with anti-pSTAT5 antibody, and analyzed by flow cytometry.

MDK-703 Induces Proliferation in PBMCs

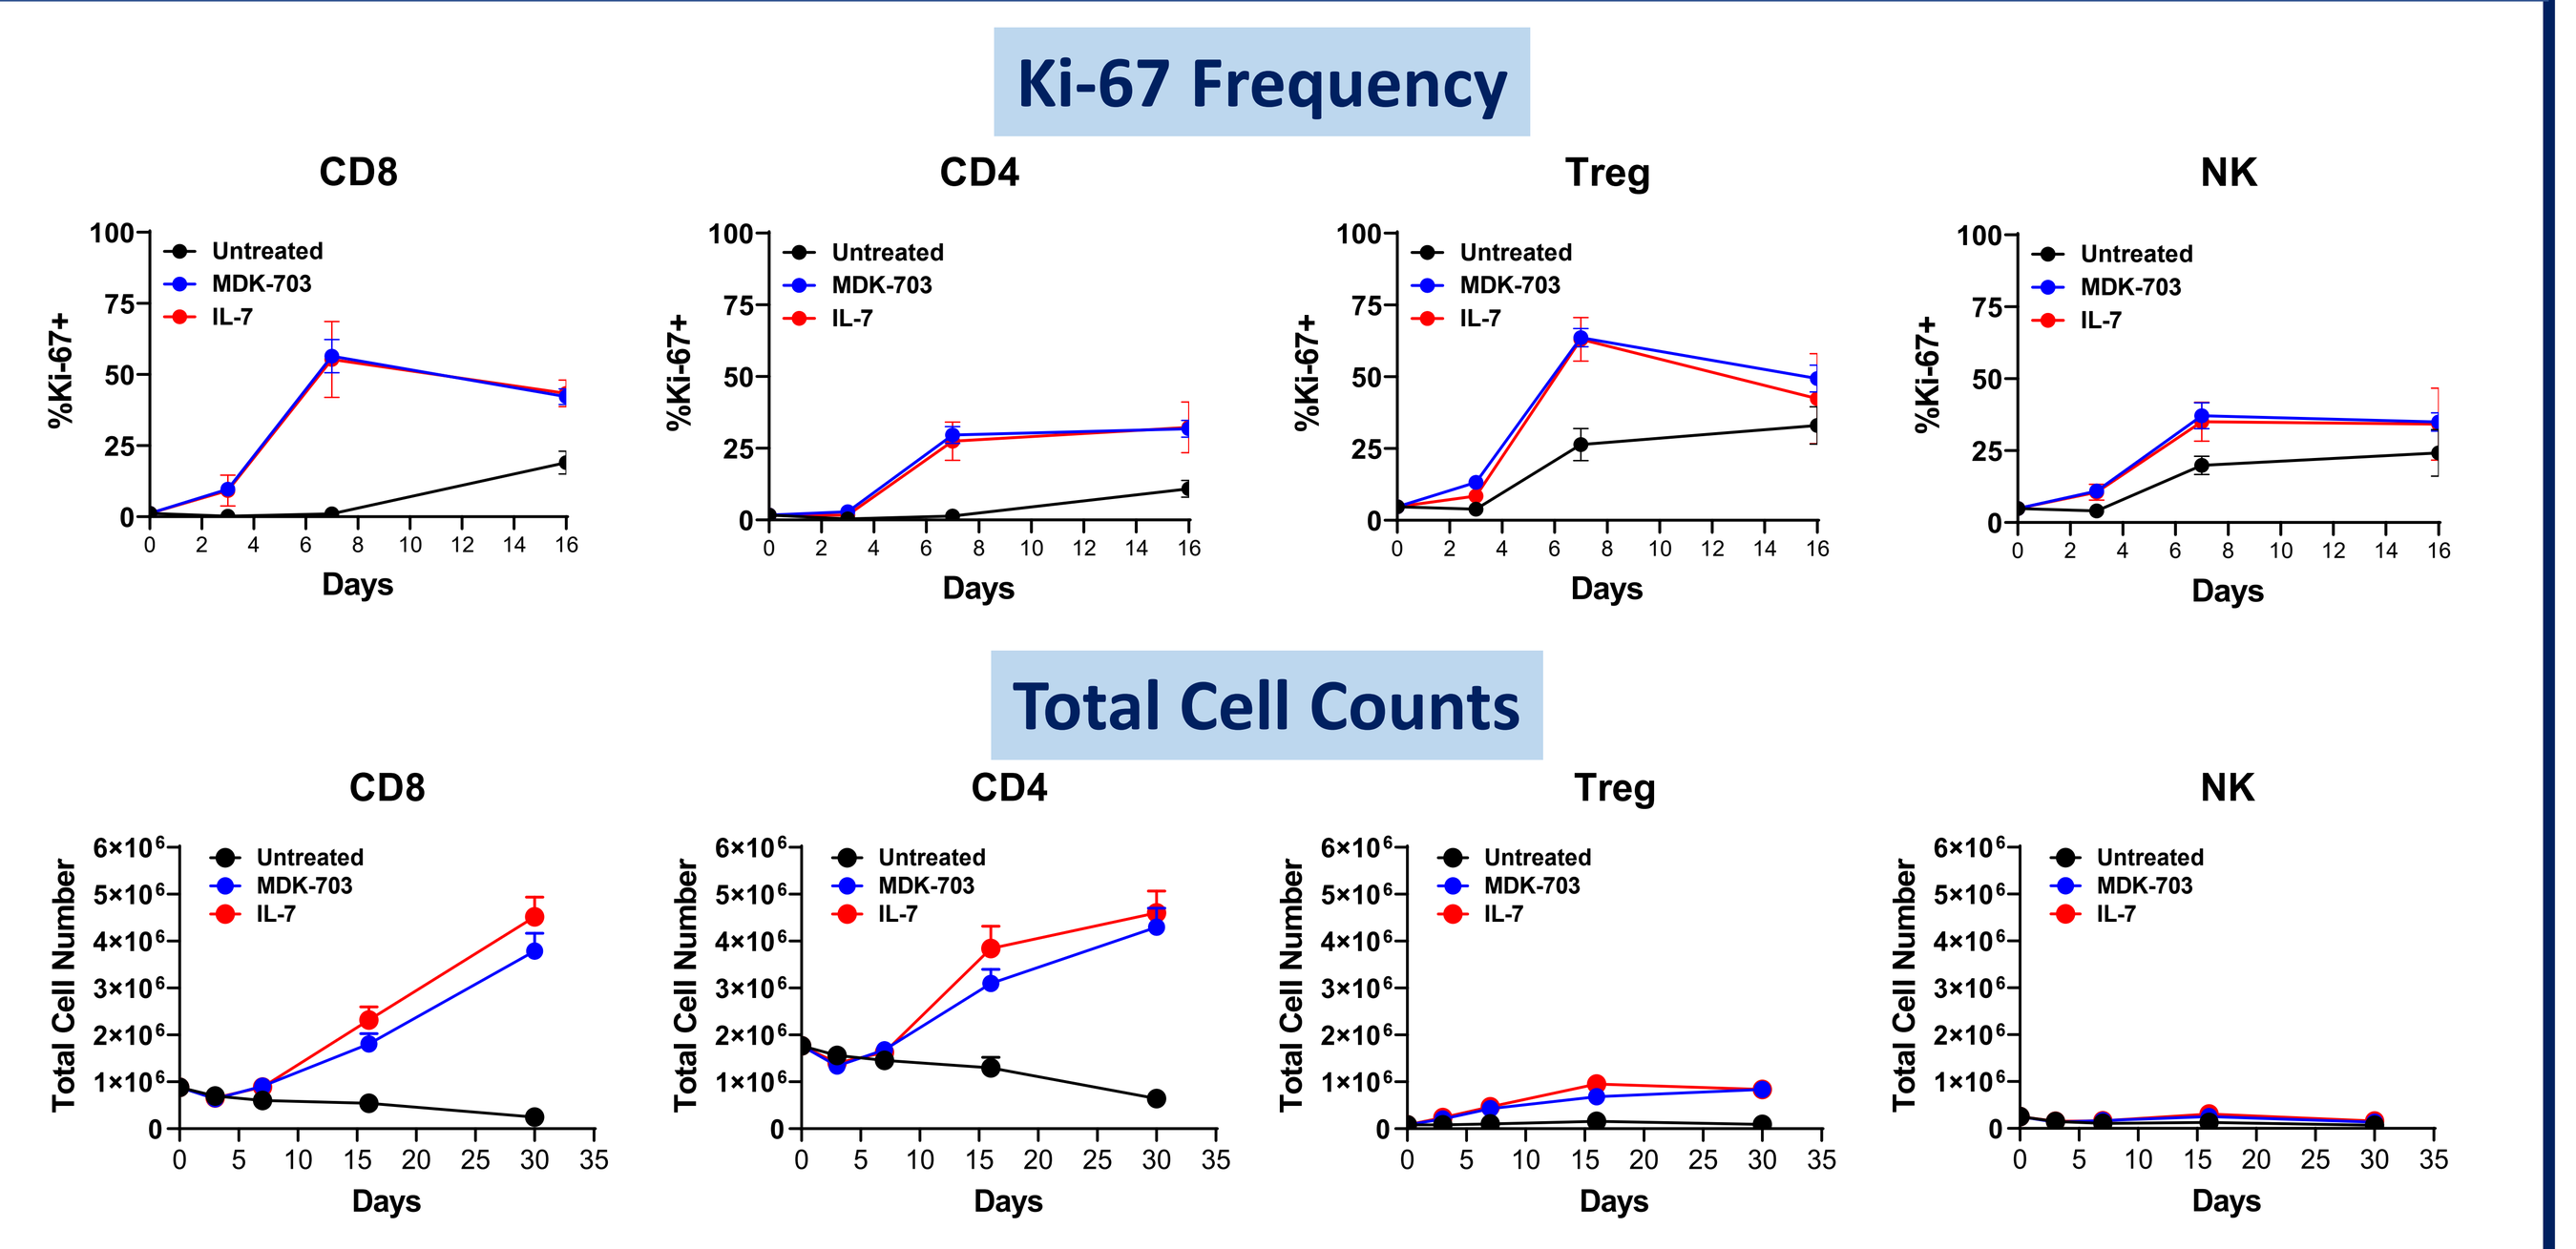
Ki-67 Frequency
CD8
CD4
Treg
NK
Untreated
MDK-703
IL-7
%Ki-67+
Days
Total Cell Counts
Total Cell Number

Fig 3. Proliferation of PBMC subpopulations following treatment with MDK-703 or IL-7. Frozen PBMCs from 5 healthy donors were rested overnight and left untreated or treated with 100nM MDK-703 or 1nM IL-7 in culture. On days 3, 7, 16, and 30, cell aliquots were taken to determine Ki-67 expression in immune cells and total cell counts in the culture.

MDK-703 Expands Tscm in PBMCs

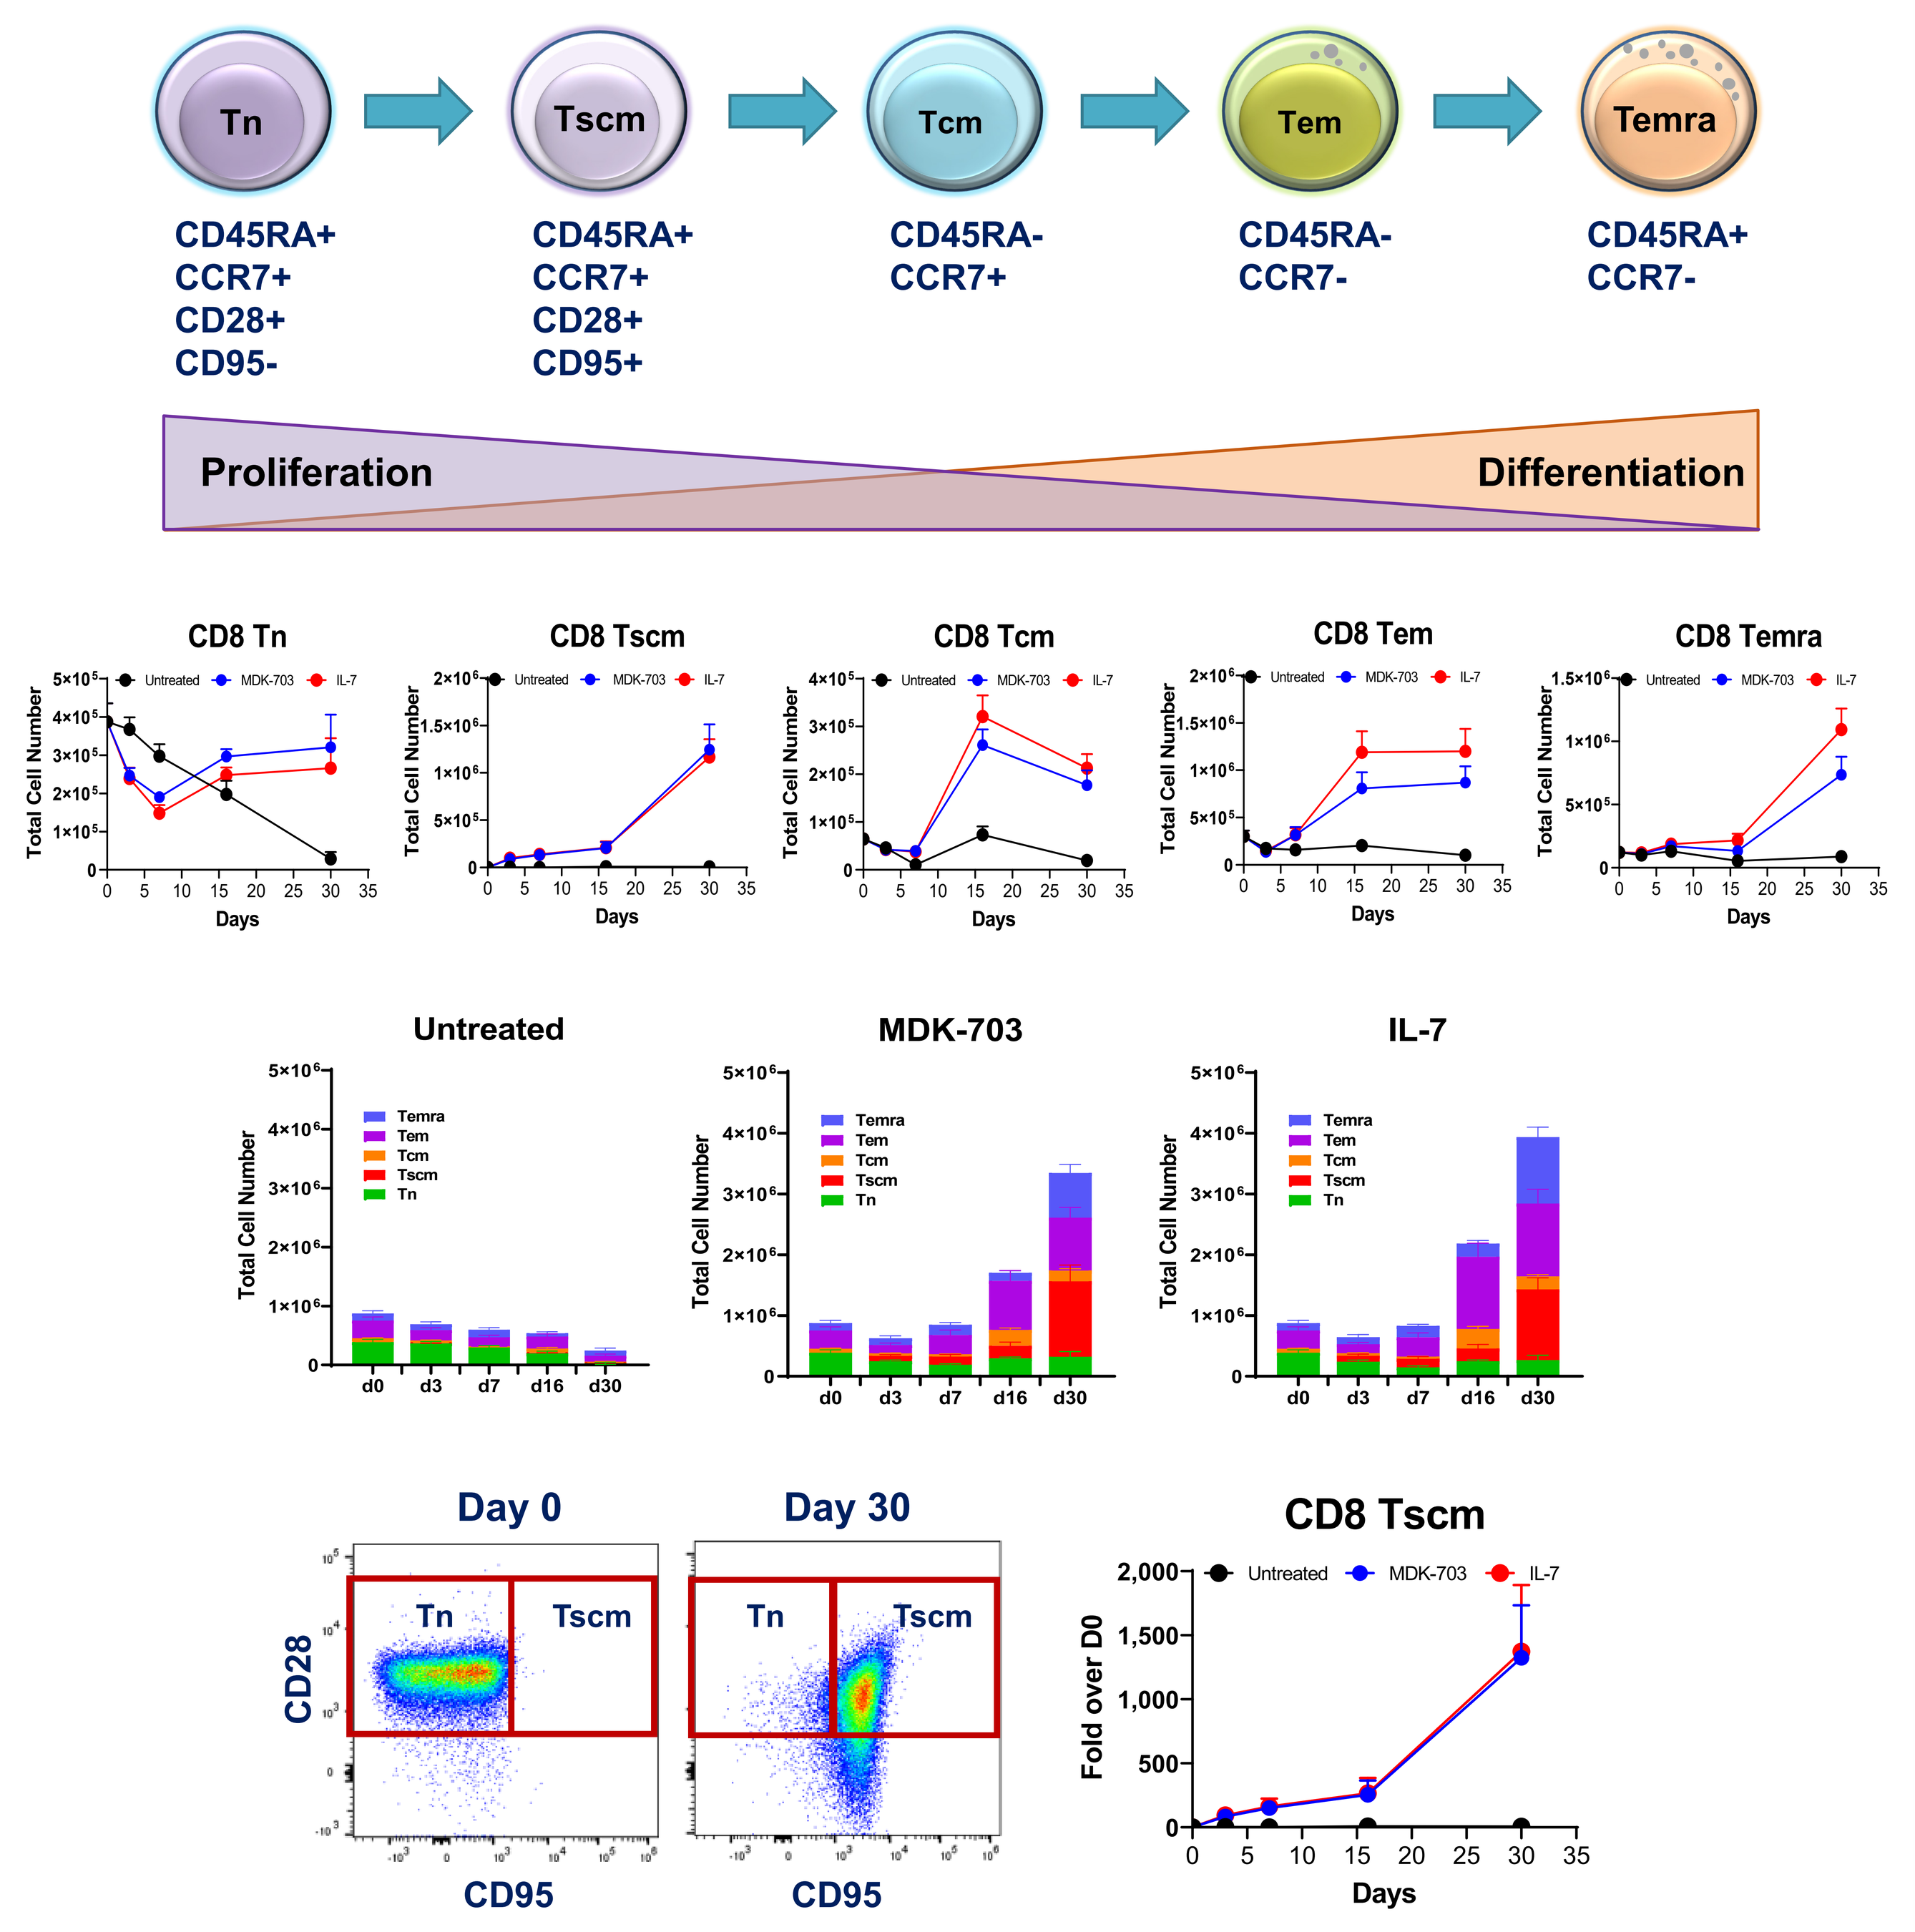
Tn
CD45RA+
CCR7+
CD28+
CD95-
Tscm
CD45RA+
CCR7+
CD28+
CD95+
Tcm
CD45RA-
CCR7+
Tem
CD45RA-
CCR7-
Temra
CD45RA+
CCR7-
Proliferation
Differentiation
CD8 Tn
CD8 Tscm
CD8 Tcm
CD8 Tem
CD8 Temra
Untreated
MDK-703
IL-7
Day 0
Day 30
CD28
CD95
CD8 Tscm
Fold over D0
Days

Fig 4. Changes in memory T-cell populations mediated by MDK-703 or IL-7 in PBMCs. Frozen PBMCs from 5 healthy donors were rested overnight and treated with vehicle, 100nM MDK-703 or 1nM IL-7 in a serum-free culture medium. On days 3, 7, 16, and 30, cell aliquots were analyzed by flow cytometry for memory T cell populations and cell counts.

MDK-703 Increases CD8, CD4, and memory T cells in Humanized Mice

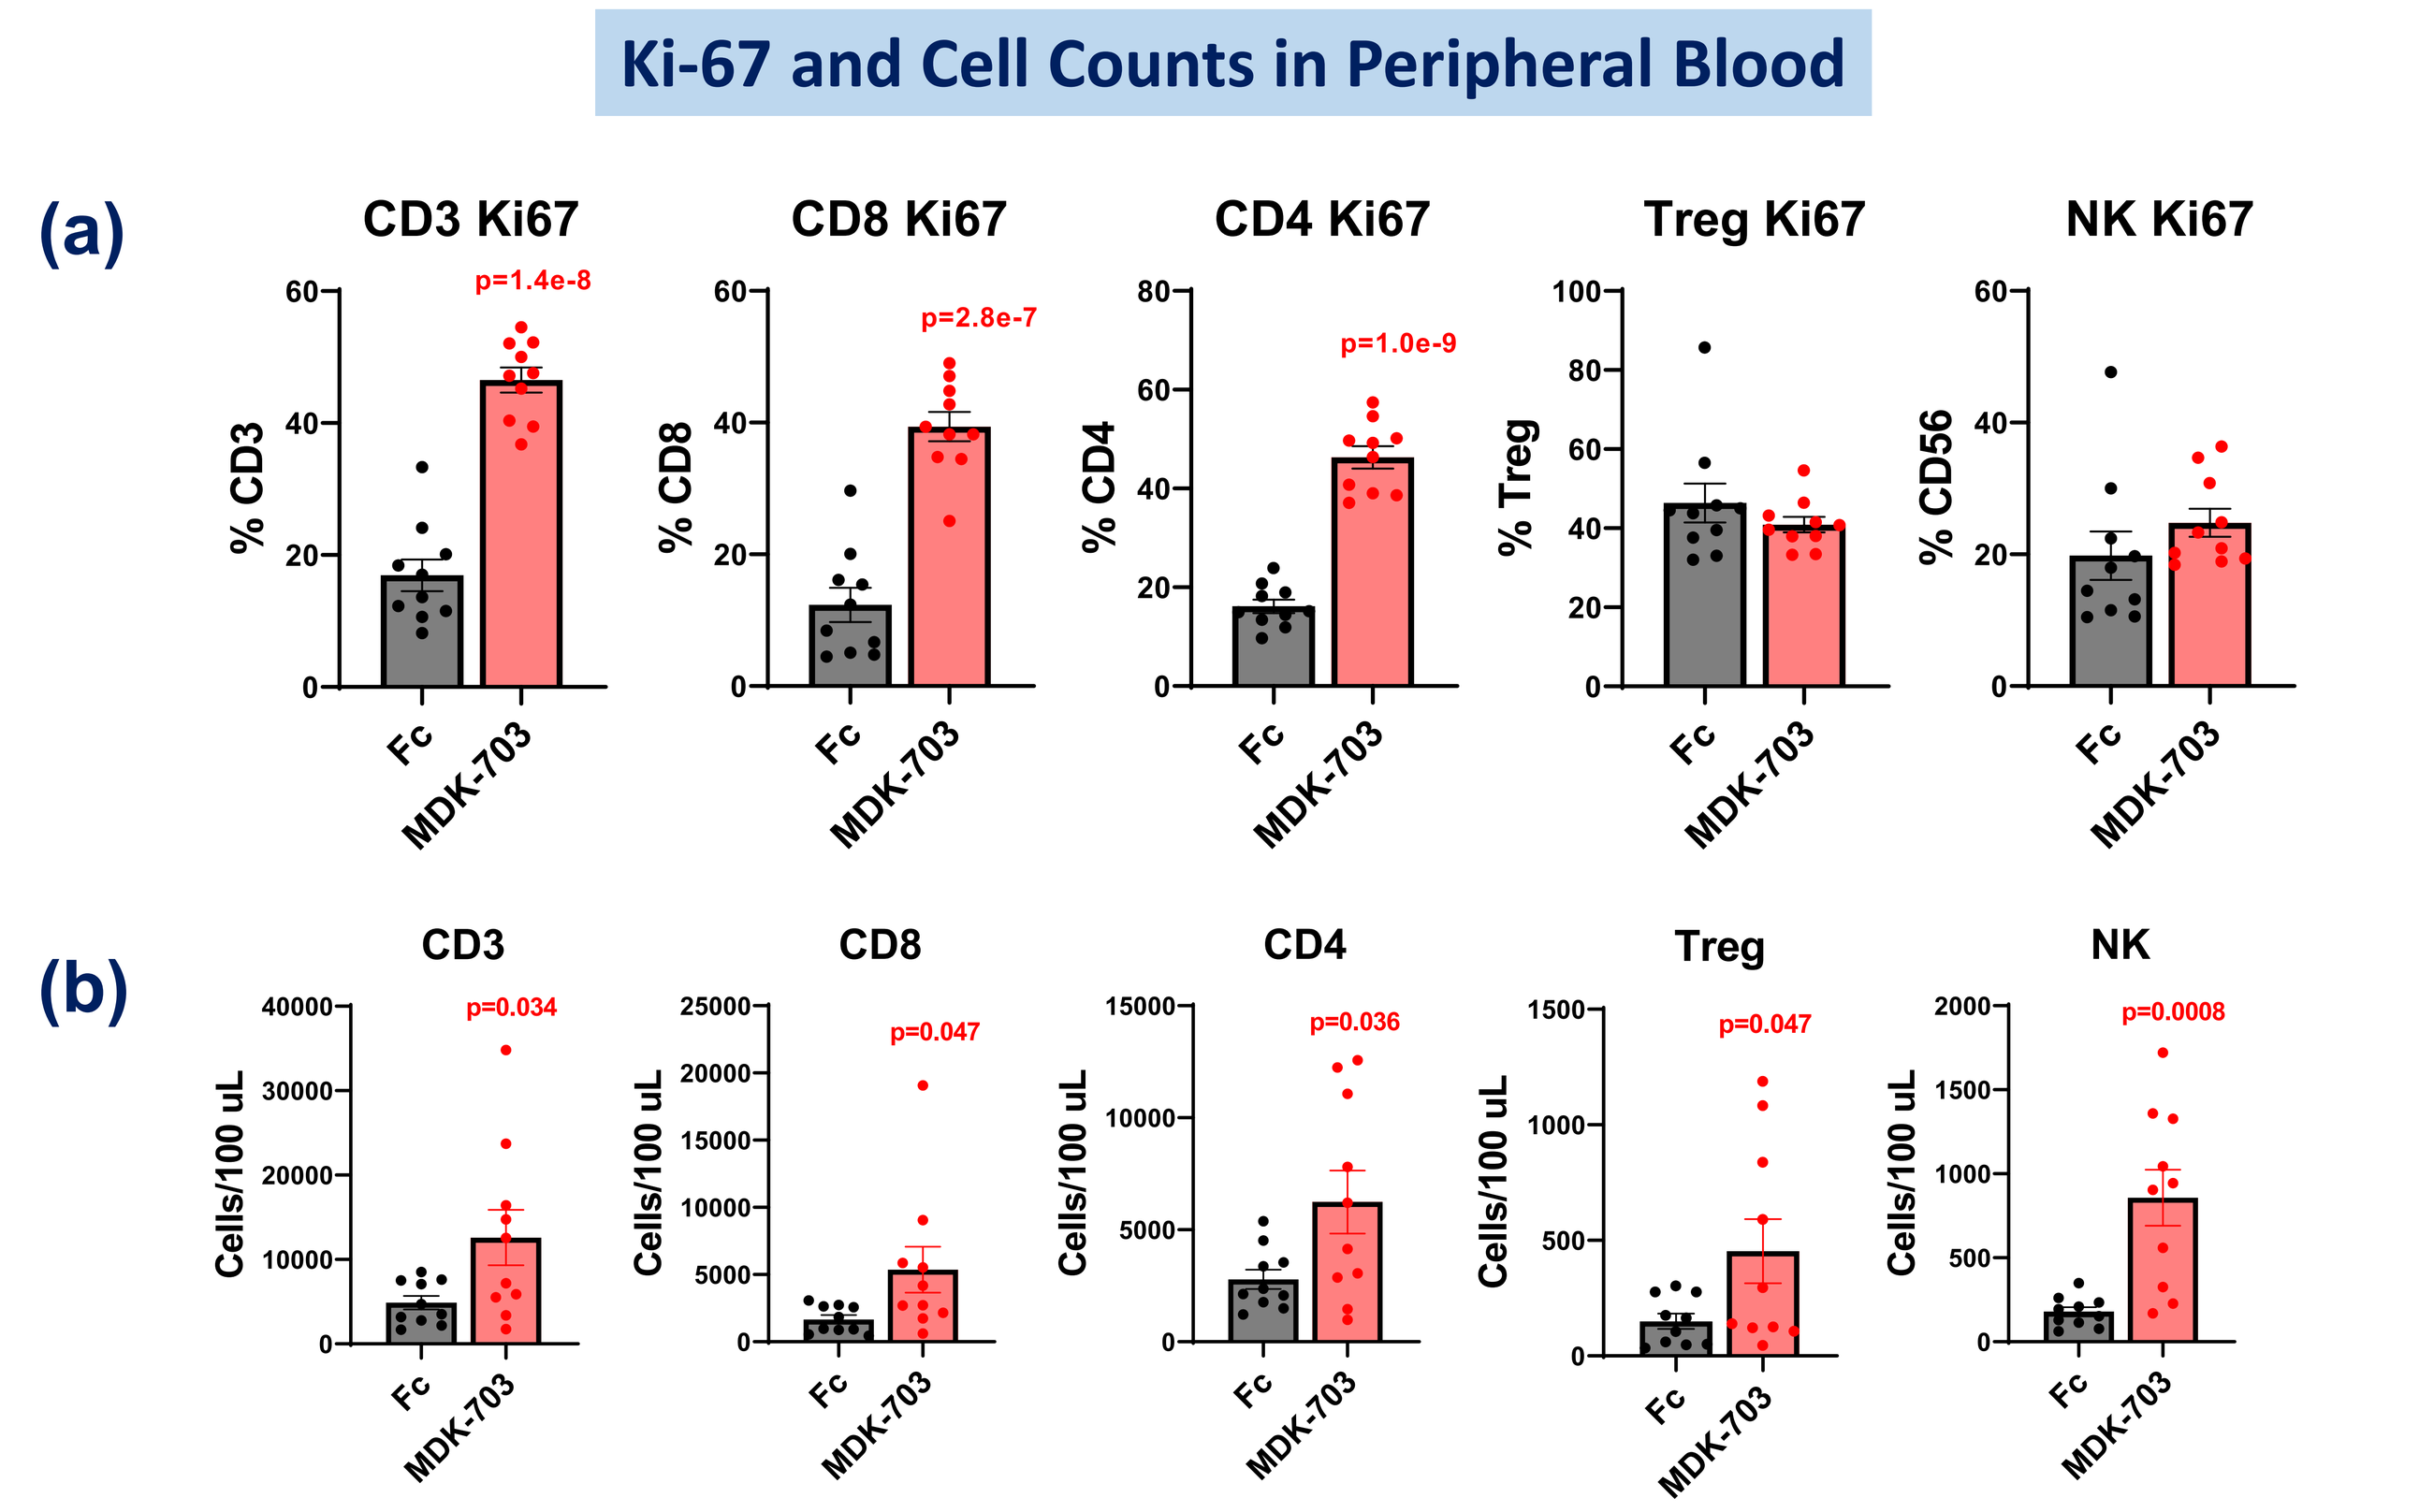
Ki-67 and Cell Counts in Peripheral Blood
(a)
CD3 Ki67
p=1.4e-8
CD8 Ki67
p=2.8e-7
CD4 Ki67
p=1.0e-9
Treg Ki67
NK Ki67
(b)
CD3
p=0.034
CD8
p=0.047
CD4
p=0.036
Treg
p=0.047
NK
p=0.0008
Fc
MDK-703

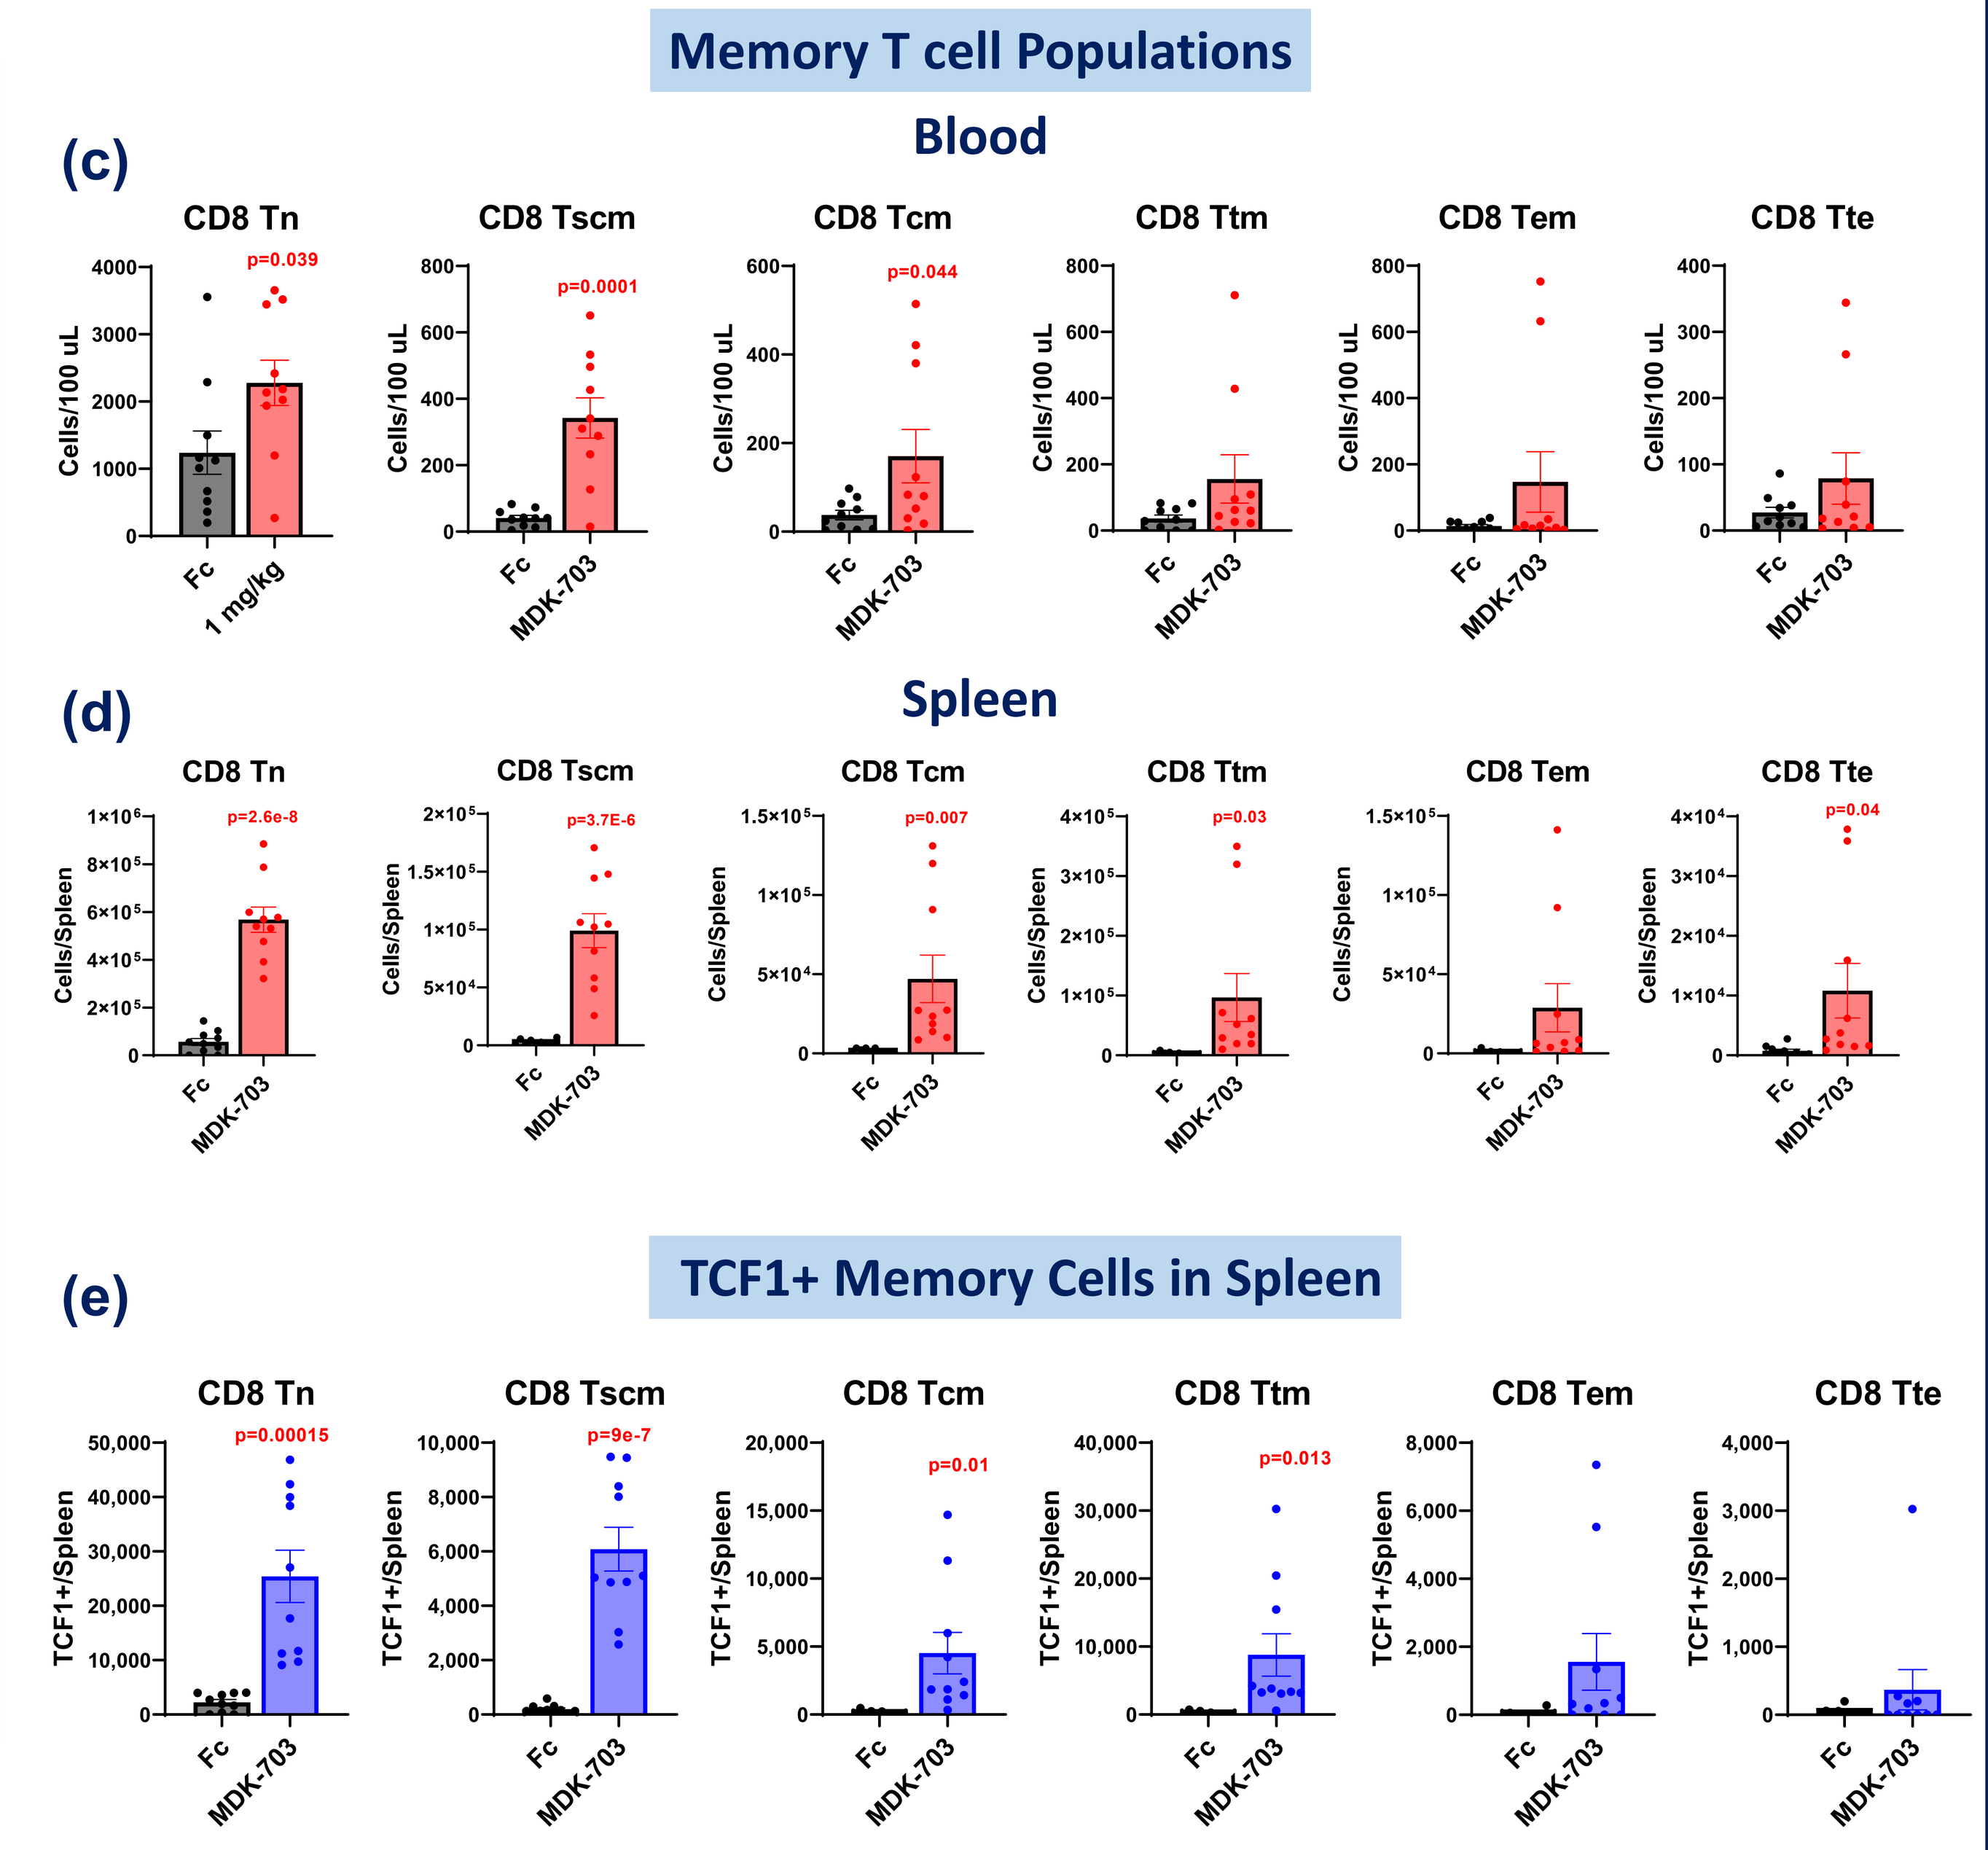
Memory T cell Populations
Blood
(c)
CD8 Tn
p=0.039
CD8 Tscm
p=0.0001
CD8 Tcm
p=0.044
CD8 Ttm
CD8 Tem
CD8 Tte
Spleen
(d)
CD8 Tn
p=2.6e-8
CD8 Tscm
p=3.7E-6
CD8 Tcm
p=0.007
CD8 Ttm
p=0.03
CD8 Tem
CD8 Tte
p=0.04
TCF1+ Memory Cells in Spleen
(e)
CD8 Tn
p=0.00015
CD8 Tscm
p=9e-7
CD8 Tcm
p=0.01
CD8 Ttm
p=0.013
CD8 Tem
CD8 Tte

Fig 5. T-cell subpopulations in humanized mice treated with MDK-703. NSG mice separately engrafted with human CD34+ cells from two donors were dosed once intravenously with 1 mg/kg Fc or MDK-703. On day 7, peripheral blood was collected to determine Ki-67+ cells in immune populations (**a**). On day 12, terminal blood samples and spleens were collected for immune profiling (**b**). Markers used for memory T cell populations (**c** and **d**) are shown in the inset box. TCF1 expression (**e**) in memory cells was also determined. Population gates were drawn based on FMO controls. Statistical analysis was done using Student's T-Test.

Tn:	CD45RA+ CCR7+ CD28+ CD95-
Tscm:	CD45RA+ CCR7+ CD28+ CD95+
Tcm:	CD45RA- CCR7+ CD28+ CD95+
Ttm:	CD45RA- CCR7- CD28+ CD95+
Tem:	CD45RA- CCR7- CD28- CD95+
Tte:	CD45RA+ CCR7- CD28- CD95+

MDK-703 PK/PD in NHP

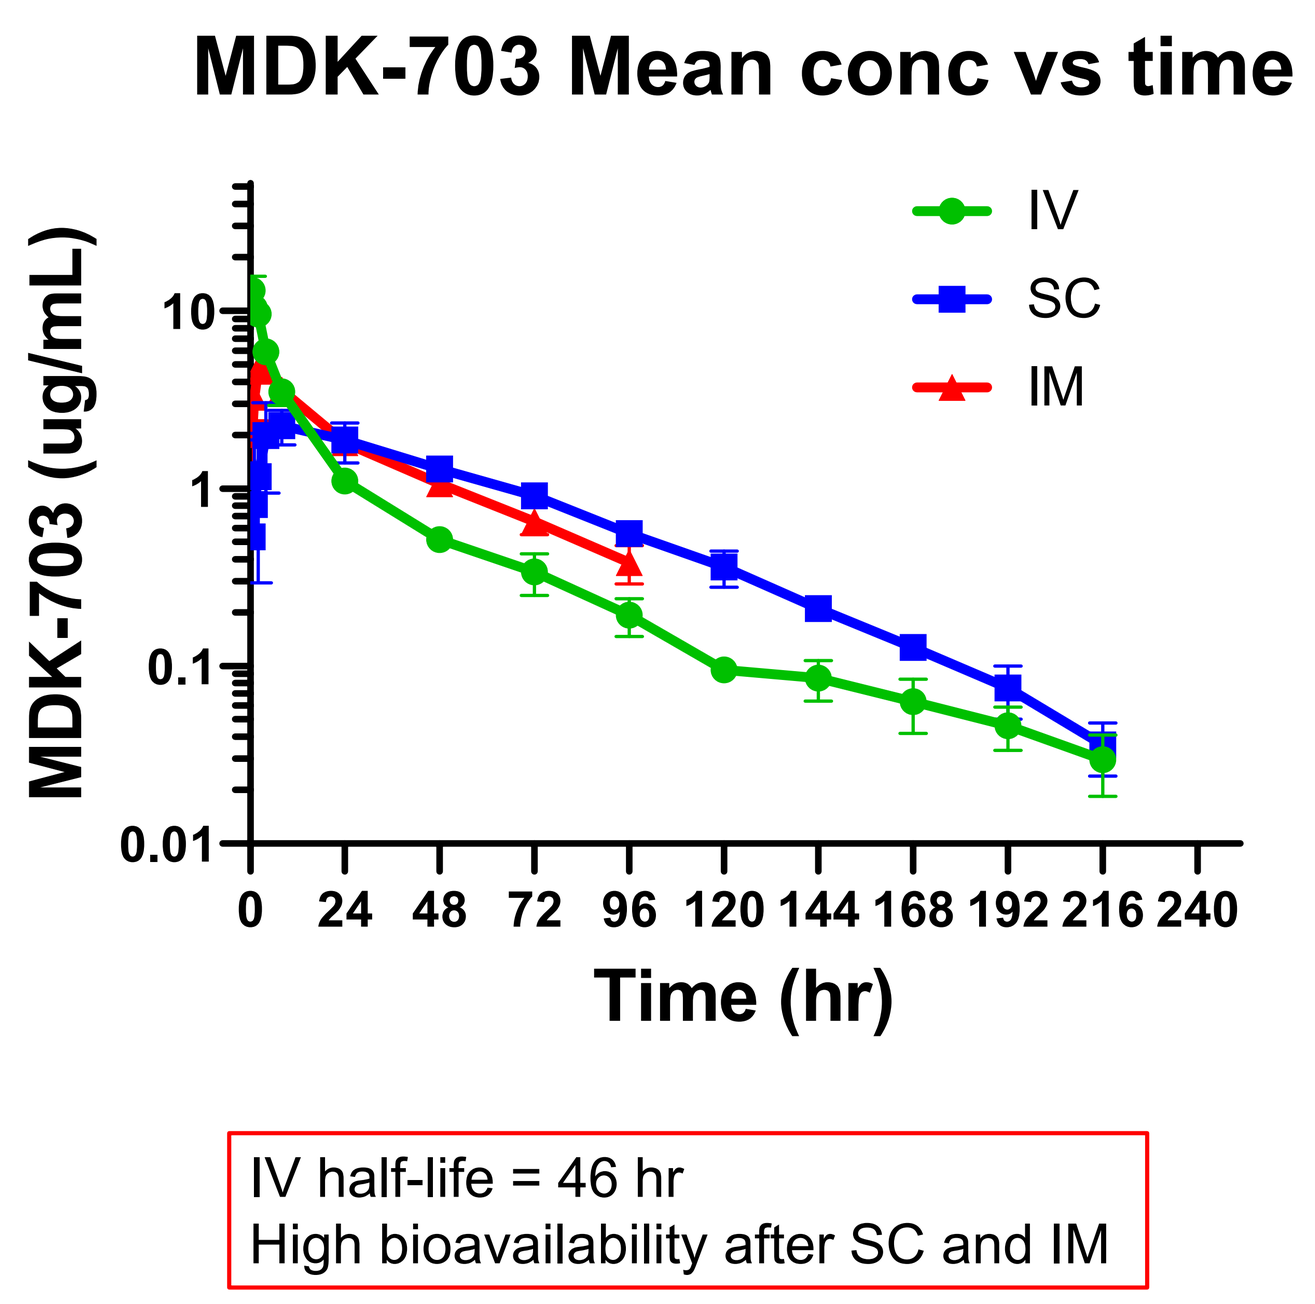
MDK-703 Mean conc vs time
IV
SC
IM
MDK-703 (ug/mL)
Time (hr)
IV half-life = 46 hr
High bioavailability after SC and IM

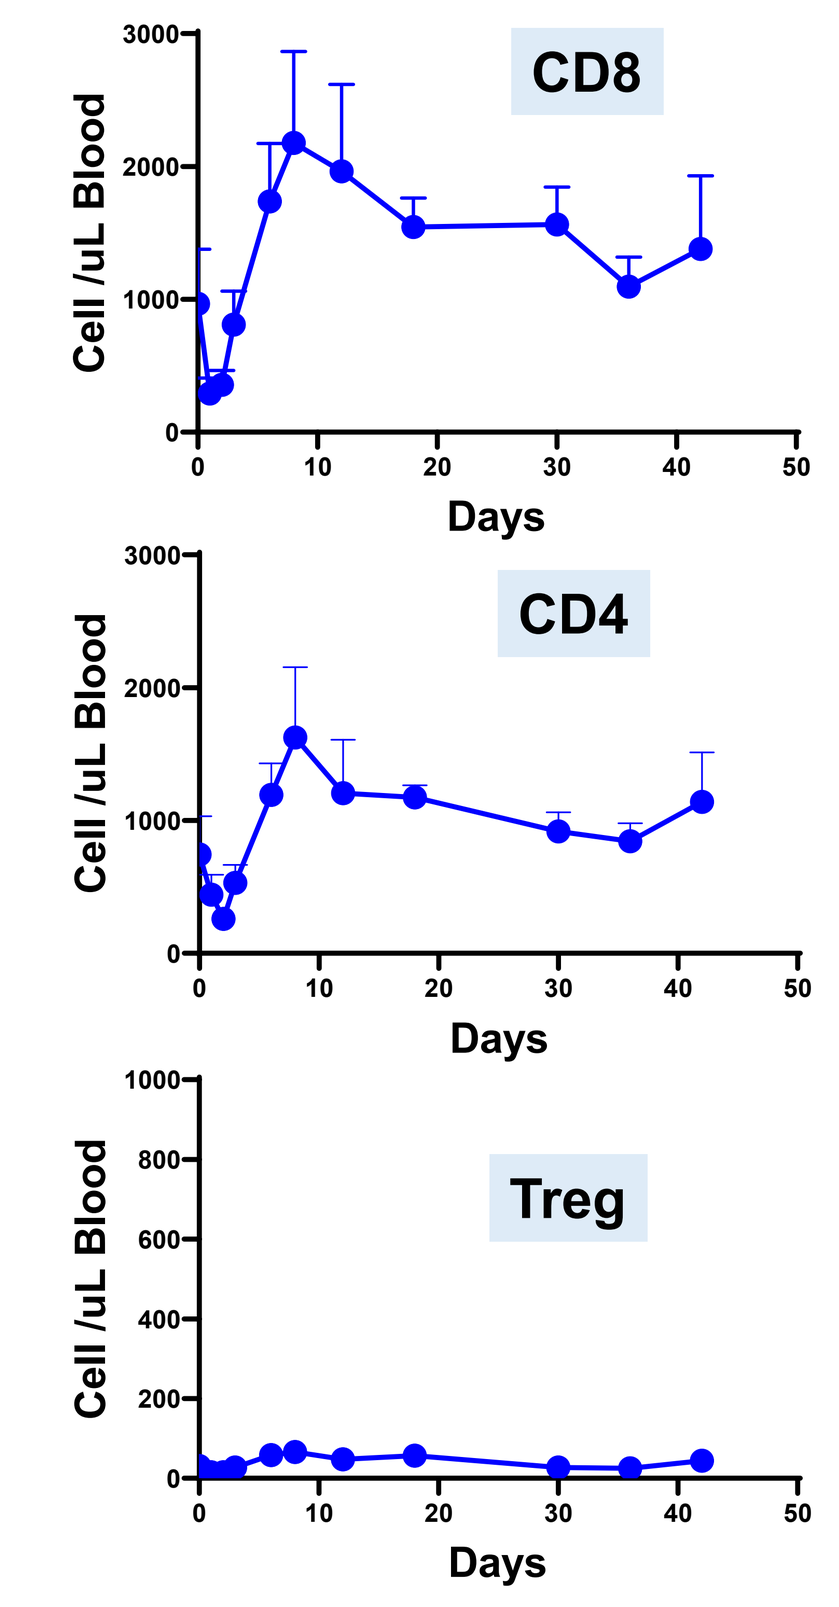
CD8
CD4
Treg
Cell /uL Blood
Days

Fig 6. Pharmacokinetics (PK) of MDK-703 in cynomolgus macaques. Animals (n=3) were administered a single dose of MDK-703 via IV, SC, or IM at 1mg/kg. The serum concentration of MDK-703 at the indicated time points was determined by sandwich ELISA.

Fig 7. Pharmacodynamic (PD) effects of MDK-703 in cynomolgus macaques. Three animals were dosed once subcutaneously at 0.3 mg/kg, and blood samples were collected at the indicated time points for immune profiling by flow cytometry.

Summary of Results

- MDK-703 binds to and activates human and NHP IL-7 receptors and emulates IL-7 in pSTAT5 assays in vitro
- In human PBMCs, MDK-703 induces T-cell differentiation and proliferation, resulting in substantial increases in memory T cell populations, including T memory stem cell (Tscm) population
- In human $CD34^+$ engrafted NSG (humanized) mice, MDK-703 increases Ki-67 expression and cell number in CD8 and CD4 cells. MDK-703 drove the expansion of memory cell populations and TCF1+ memory cells in both blood and spleen on day 12
- In cynomolgus macaques, MDK-703 exhibited a $T_{1/2}$ of 46 hours after IV administration and showed high bioavailability after IM and SC dosing. A single subcutaneous dose at 300 ug/kg induced a sustained increase in $CD8^+$ and $CD4^+$ T-cells for up to 40 days

Conclusions

MDK-703 is a full agonist of the IL-7 receptor and expanded CD8, CD4, and memory T cell populations in PBMC cultures and in vivo in a humanized mouse model. Especially noteworthy was the expansion of TCF1+ stem-like memory cells which have been shown to possess self-renewal potential.

MDK-703 offers an attractive alternative to IL-7-based drug candidates currently in clinical development. The structural novelty of MDK-703 eliminates the risk of ADAs that neutralize endogenous IL-7. A first human trial of MDK-703 is anticipated in 2022.

References

1) Clin Transl Sci (2020) 13, 1161–1169
2) JITC-2020-SITC2020.0567
3) Human Vaccines & Immunotherapeutics (2015) 11, 2312-2321